Simplified test and treat protocols for population level screening and elimination of hepatitis B and hepatitis C in Uzbekistan

E. MUSABAEV[1], S. SADIROVA[1], S. BAKIEVA[1], K. BRIGIDA[1], R. DUNN[2], K. RAZAVI-SHEARER[2], H. RAZAVI[2]

1 Uzbekistan Research Institute of Virology

2 Center for Disease Analysis Foundation

INTRODUCTION

In July of 2017, in the Republic of Uzbekistan, the Cabinet of Ministers signed a Decree to provide additional measures to prevent the spread of infectious diseases. This expansive decree specifically addresses the need to gradually increase the diagnosis of hepatitis B virus (HBV) and hepatitis C virus (HCV).

In partnership with the Uzbekistan Research Institute of Virology (RIV), CDAF implemented a pilot project – Uzbekistan Hepatitis Elimination Program (UHEP) – starting on December 6, 2019 and concluding on December 31, 2020. The program utilized decentralized testing, simplified diagnostic algorithms and treatment of uncomplicated cases by general practitioners.

The program was paused between March 15th and August 25th due to the COVID pandemic. Operations resumed in late August with simultaneous screening of patients for HBV, HCV and SARS-CoV-2 (COVID-19).

AIMS

An estimated 3.5 million people are infected with HBV or HCV in Uzbekistan1. This study tested the feasibility of eliminating HBV & HCV if the testing and treatment protocols were simplified to support a national program.

METHOD

The study was conducted in the capital city of Tashkent over a 12 month period with 6 months of inactivity due to COVID-19. Screening and onsite follow-up testing took place at multiple polyclinics located in Tashkent.

SIMPLIFIED TEST & TREAT: Simplified testing algorithms were developed by a medical advisory board and approved by the Uzbekistan Ministry of Health (MOH).

COST CONTROL: Tests and medicines were procured at low prices by the Global Procurement Fund (GPRO) [1]. The government waived most import duties and fees and provided human resources, clinic space, and laboratory equipment and supplies. A national pharmacy chain was contracted to sell medicines at only a 5% mark-up.

DATA COLLECTION: A REDCap patient registry was customized and used to record patients' consent, contact information, medical history, test results, and doctors' notes using low cost handheld tablets.

TRAINING: Nurses were trained to use rapid HCV antibody and hepatitis B surface antigen (HBsAg) to screen patients at polyclinics. Rapid creatinine and human immunodeficiency virus (HIV) tests were used to test HBsAg+ patients before they were referred for treatment.

TESTING: All patients received testing for HBsAg and anti-HCV rapid tests. HBsAg+ patients were immediately tested for HIV and creatinine levels. Anti-HCV+ patients immediately had blood drawn for off-site laboratory analysis of HCV core antigen, creatinine, aspartate aminotransferase (AST), and platelets.

ELIGIBILITY and TREATMENT:

- HBV: All patients with a positive HBsAg test, a negative HIV test and normal renal function (estimated glomerular filtration rate [eGFR] >50 mL/min/1.73 m2) were determined to be eligible for treatment for hepatitis B infection and were offered a 12-month prescription for tenofovir disoproxil fumarate with instructions to return after 12 months for free follow-up tests (HBsAg, HIV, and creatinine). All HIV-positive patients were referred to HIV clinics for treatment.
- HCV: Cirrhotic patients (APRI>1.5) and patients with abnormal renal function (eGFR <30 mL/min/1.73 m2) were referred to the RIV for treatment outside of the program. All other HCV core antigen-positive patients were considered eligible for treatment and offered a 3-month prescription for sofosbuvir/daclatasvir and instructed to return after 12 weeks following completion of treatment for a free SVR test.

TASK-SHARING: For the first 3 months of the program, patients were treated by liver specialists of the RIV. For the remainder of the program, patients with uncomplicated cases were treated by trained general practitioners at their local polyclinic where screening was conducted while cirrhotic patients were referred to the RIV liver specialists for treatment.

TREATMENT COSTS: 90% of patients paid for treatment at the negotiated prices: $180 for 12 months of tenofovir disoproxil fumarate (TDF); $204 for a 3-month treatment of Sofosbuvir/Daclatasvir (SOF/DAC). 10% of patients who were too poor to pay for treatment received free treatment.

RESULTS

More that 60,000 people were screened at polyclinics in 6 months of operation in spite of lower patient engagement with healthcare systems due to COVID-19. 69% of HBV+ were linked to care. 73% of those patients received a prescription, of which 65% initiated treatment. 54% of HCV+ were linked to care. 74% of viremic patients received a prescription, of which 56% initiated treatment. There were large geographic variations in HBV and HCV prevalence across Tashkent and nationally.

Figure 1. HBV and HCV Cascade of Care

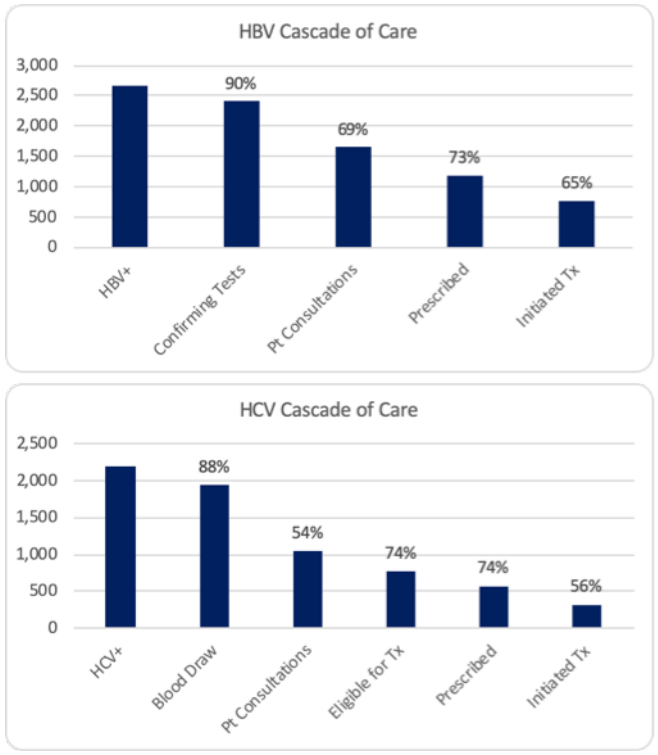
HBV Cascade of Care
3,000
2,500
2,000
1,500
1,000
500
0
90%
69%
73%
65%
HBV+
Confirming Tests
Pt Consultations
Prescribed
Initiated Tx
HCV Cascade of Care
2,500
2,000
1,500
1,000
500
0
88%
54%
74%
74%
56%
HCV+
Blood Draw
Pt Consultations
Eligible for Tx
Prescribed
Initiated Tx

Figure 2. Treatment Discontinuation Rates

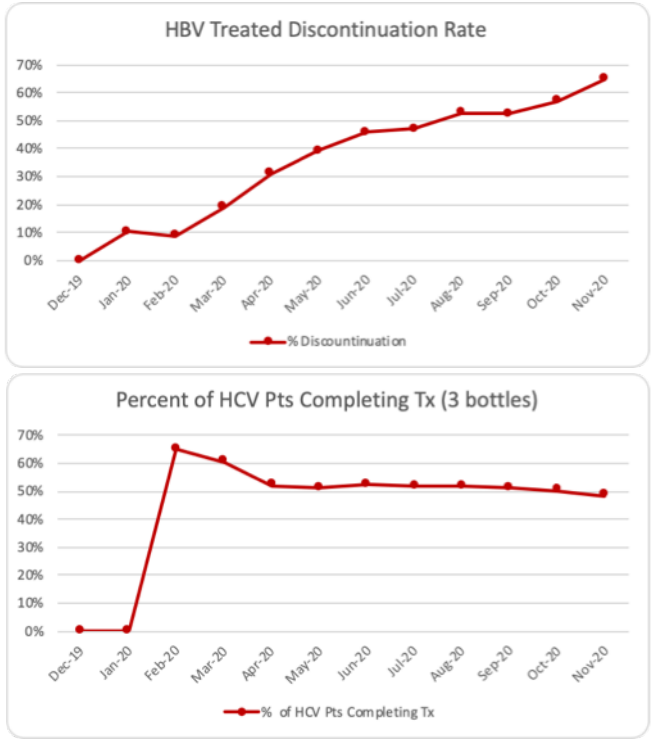
HBV Treated Discontinuation Rate
70%
60%
50%
40%
30%
20%
10%
0%
Dec-19
Jan-20
Feb-20
Mar-20
Apr-20
May-20
Jun-20
Jul-20
Aug-20
Sep-20
Oct-20
Nov-20
% Discountinuation
Percent of HCV Pts Completing Tx (3 bottles)
70%
60%
50%
40%
30%
20%
10%
0%
Dec-19
Jan-20
Feb-20
Mar-20
Apr-20
May-20
Jun-20
Jul-20
Aug-20
Sep-20
Oct-20
Nov-20
% of HCV Pts Completing Tx

Figure 3. Hepatitis Prevalence by Region of Birth and Residence

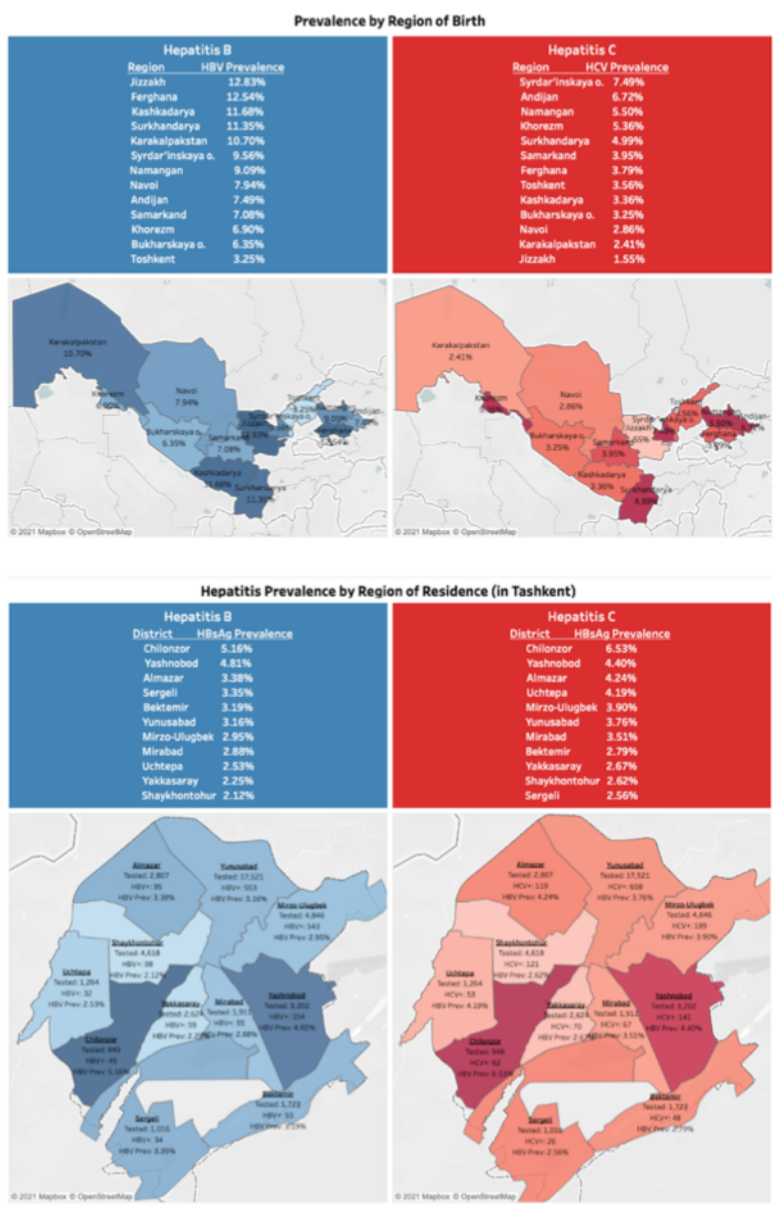
Prevalence by Region of Birth
Hepatitis B
Region HBV Prevalence
Jizzakh 12.83%
Ferghana 12.54%
Kashkadarya 11.68%
Surkhandarya 11.35%
Karakalpakstan 10.70%
Syrdar'inskaya o. 9.56%
Namangan 9.09%
Navoi 7.94%
Andijan 7.49%
Samarkand 7.08%
Khorezm 6.90%
Bukharskaya o. 6.35%
Toshkent 3.25%
Hepatitis C
Region HCV Prevalence
Syrdar'inskaya o. 7.49%
Andijan 6.72%
Namangan 5.50%
Khorezm 5.36%
Surkhandarya 4.99%
Samarkand 3.95%
Ferghana 3.79%
Toshkent 3.56%
Kashkadarya 3.36%
Bukharskaya o. 3.25%
Navoi 2.86%
Karakalpakstan 2.41%
Jizzakh 1.55%
Hepatitis Prevalence by Region of Residence (in Tashkent)
Hepatitis B
District HBsAg Prevalence
Chilonzor 5.16%
Yashnobod 4.81%
Almazar 3.38%
Sergeli 3.35%
Bektemir 3.19%
Yunusabad 3.16%
Mirzo-Ulugbek 2.95%
Mirabad 2.88%
Uchtepa 2.53%
Yakkasaray 2.25%
Shaykhontohur 2.12%
Hepatitis C
District HBsAg Prevalence
Chilonzor 6.53%
Yashnobod 4.40%
Almazar 4.24%
Uchtepa 4.19%
Mirzo-Ulugbek 3.90%
Yunusabad 3.76%
Mirabad 3.51%
Bektemir 2.79%
Yakkasaray 2.67%
Shaykhontohur 2.62%
Sergeli 2.56%
© 2021 Mapbox © OpenStreetMap

***Figure 4.** Percentage of Eligible Patients Initiating Treatment, by Doctor*

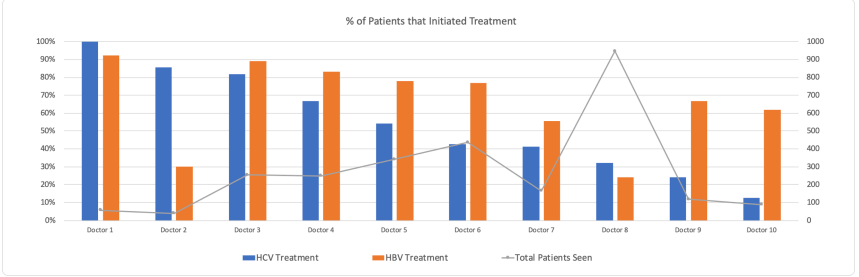
% of Patients that Initiated Treatment
HCV Treatment
HBV Treatment
Total Patients Seen

@CDAFound

CONCLUSIONS

A simplified test/treat protocol can be used to screen, link to care and treat large numbers of HCV+ and HBsAg+ patients using nurses and GPs who are accessible throughout the country. Patients with advanced liver disease still require consultation with a liver specialist.

Mass screening for hepatitis can be successfully conducted when simultaneously screening for SARS-CoV-2 (COVID-19).

Use of pooled procurement and negotiated pricing enabled patients in a middle-income country to afford treatment. This approach may be used to offset overall elimination costs in a national program.

The study highlighted hotspots in the country that could be prioritized in a national program.

Future programs would benefit from research to understand factors affecting treatment initiation rates among doctors and addition of patient navigators to reduce treatment discontinuation rates.

ACKNOWLEDGEMENTS

We would like to thank all of our collaborators with special thanks to the RIV staff who manage daily program operations.

We would also like to thank the US Embassy to Uzbekistan and Mr. Akhror Burkhanov, Cultural Attaché at the Uzbekistan Embassy to the US, for their support in navigating political processes and obtaining necessary decrees to enable this study.

We would like to thank the Dentons law firm in Tashkent for their services in understanding and complying with Uzbekistan laws.

This study was funded by the Center for Disease Analysis Foundation and through grants from the John C. Martin Foundation and Gilead Sciences. Gilead Sciences had no influence over the design, analysis, or recommendations.

REFERENCES

1 **Global Procurement Fund**, www.cdafound.org/gpro

CONTACT INFORMATION

Rick Dunn, Center for Disease Analysis Foundation
rdunn@cdafound.org